ORIC-101 Comprehensively Inhibits Glucocorticoid Pathways to Overcome Therapeutic Resistance in Pan-Cancer Models

Aleksandr Pankov, Haiying Zhou, Shravani Barkund, Ganapati Hegde, Padmini Narayanan, Omar Kabbarah, Lori S. Friedman, Anneleen Daemen
ORIC Pharmaceuticals, South San Francisco, CA 94080

BACKGROUND

The Glucocorticoid Receptor (GR) is a member of the nuclear receptor superfamily of ligand-activated transcription factors. GR can be activated both by endogenous glucocorticoid ligands, including cortisol and corticosterone, and by synthetic glucocorticoid drugs, such as dexamethasone (Dex).

Preclinical studies have established a role for GR in mediating resistance to both anti-hormonal therapies and conventional chemotherapies in epithelial cancers (Zhang et al., 2006; Arora et al., 2013; Isikbay et al., 2014; Stringer-Reasor et al., 2015).

Studies in breast cancer models have established that the chemoprotective effect afforded by GR activation is facilitated, at least partially, through upregulation of genes involved in antiapoptosis, epithelial/mesenchymal transition (EMT), and metastasis – thus suggesting that GR inhibition may enhance therapeutic responses (Skor et al., 2013; Obradović et al., 2019; Shi et al., 2019).

Opportunity: Understand molecular consequences of GR signaling and identify a patient subpopulation more likely to benefit from GR inhibition.

1. ORIC-101: a potent small molecule GR antagonist

ORIC-101 (Steroidal)
GR antagonism IC_{50} = 7.3 nM
AR agonism IC_{50} > 5000 nM
PR antagonism IC_{50} = 22 nM
CYP3A4 IC_{50} = 1.6 µM
CYP2C8/CYP2C9 IC_{50} > 10 µM

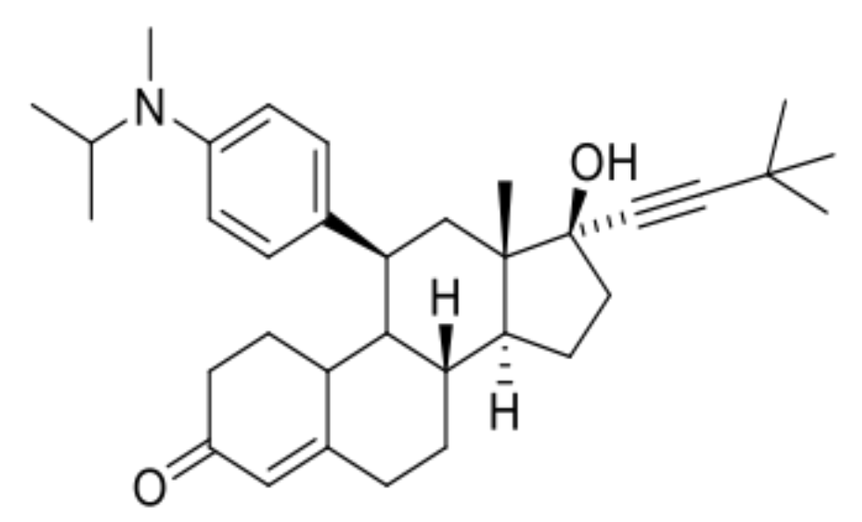

Rew et al., 2018

- ORIC-101 overcomes resistance to diverse chemotherapeutics across cancer types AACR 2020 Poster #4121
- ORIC-101 overcomes GR-mediated chemoresistance in pancreatic cancer models AACR 2020 Poster #4123

2. ORIC-101 shows modulation of common GR targets in surrogate tissues of healthy volunteers

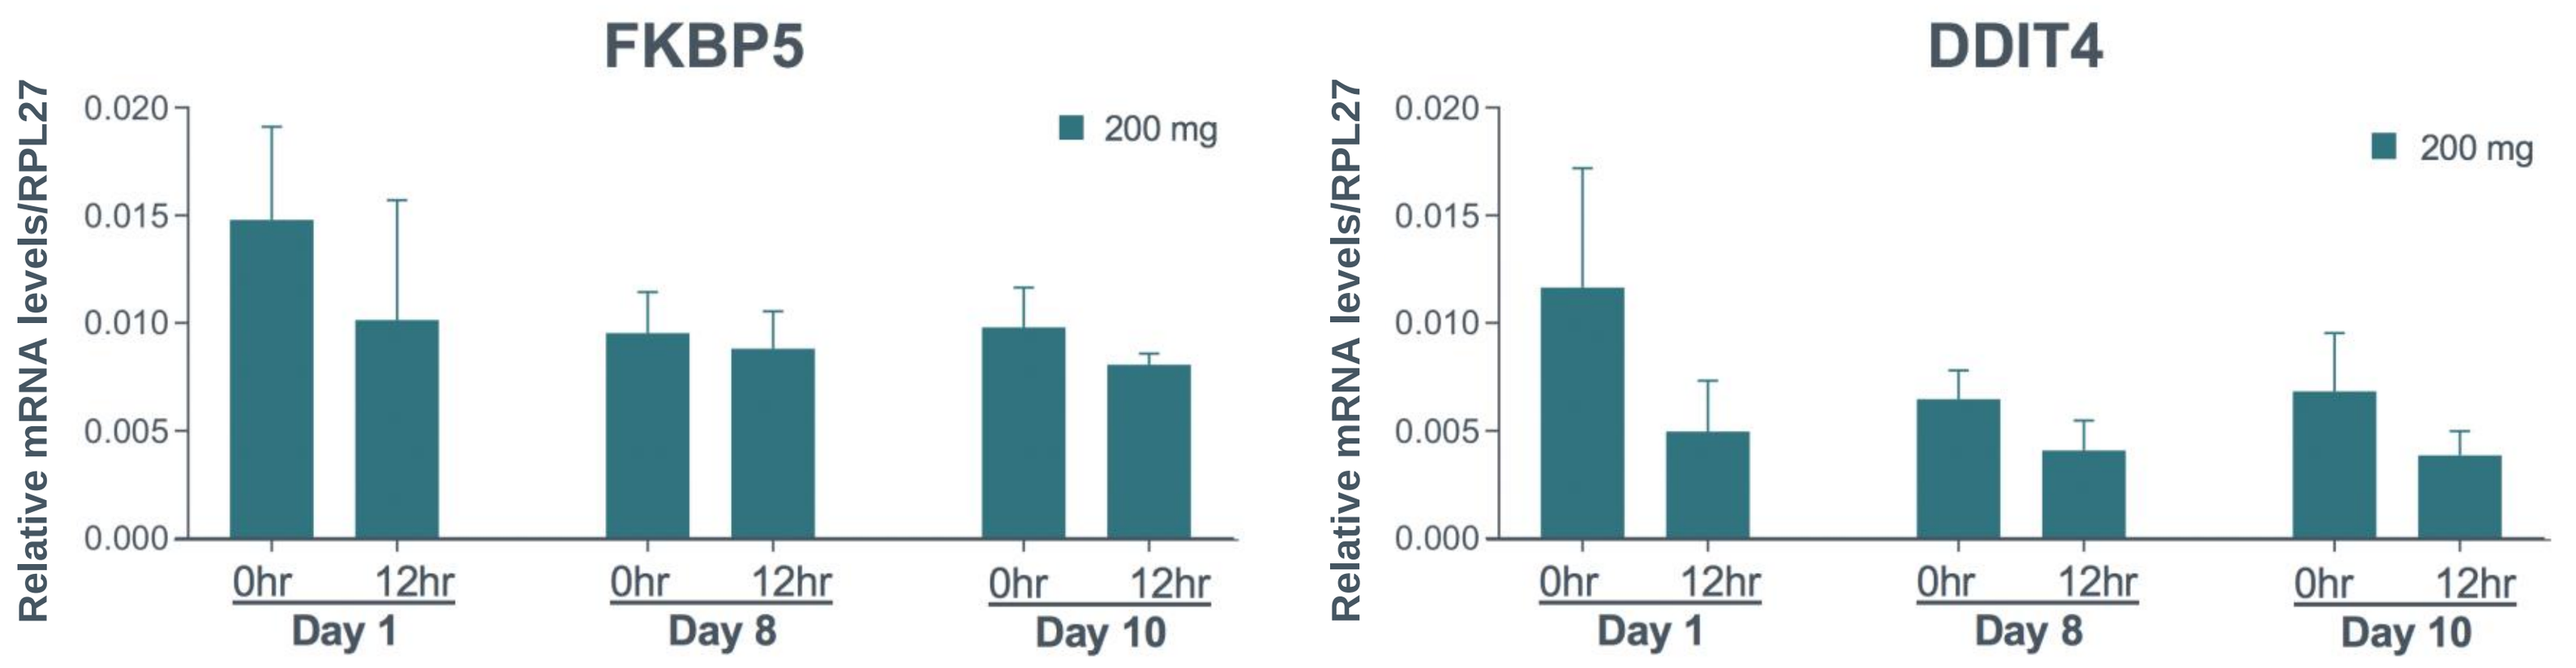
FKBP5
DDIT4
200 mg
Relative mRNA levels/RPL27
0.020
0.015
0.010
0.005
0.000
0hr
12hr
Day 1
Day 8
Day 10

Reference: Internal company data

Rapid and sustained functional GR inhibition was demonstrated in PBMCs of healthy volunteers with once-daily ORIC-101

3. Precision medicine strategy focuses on identifying patients with activated GR signaling

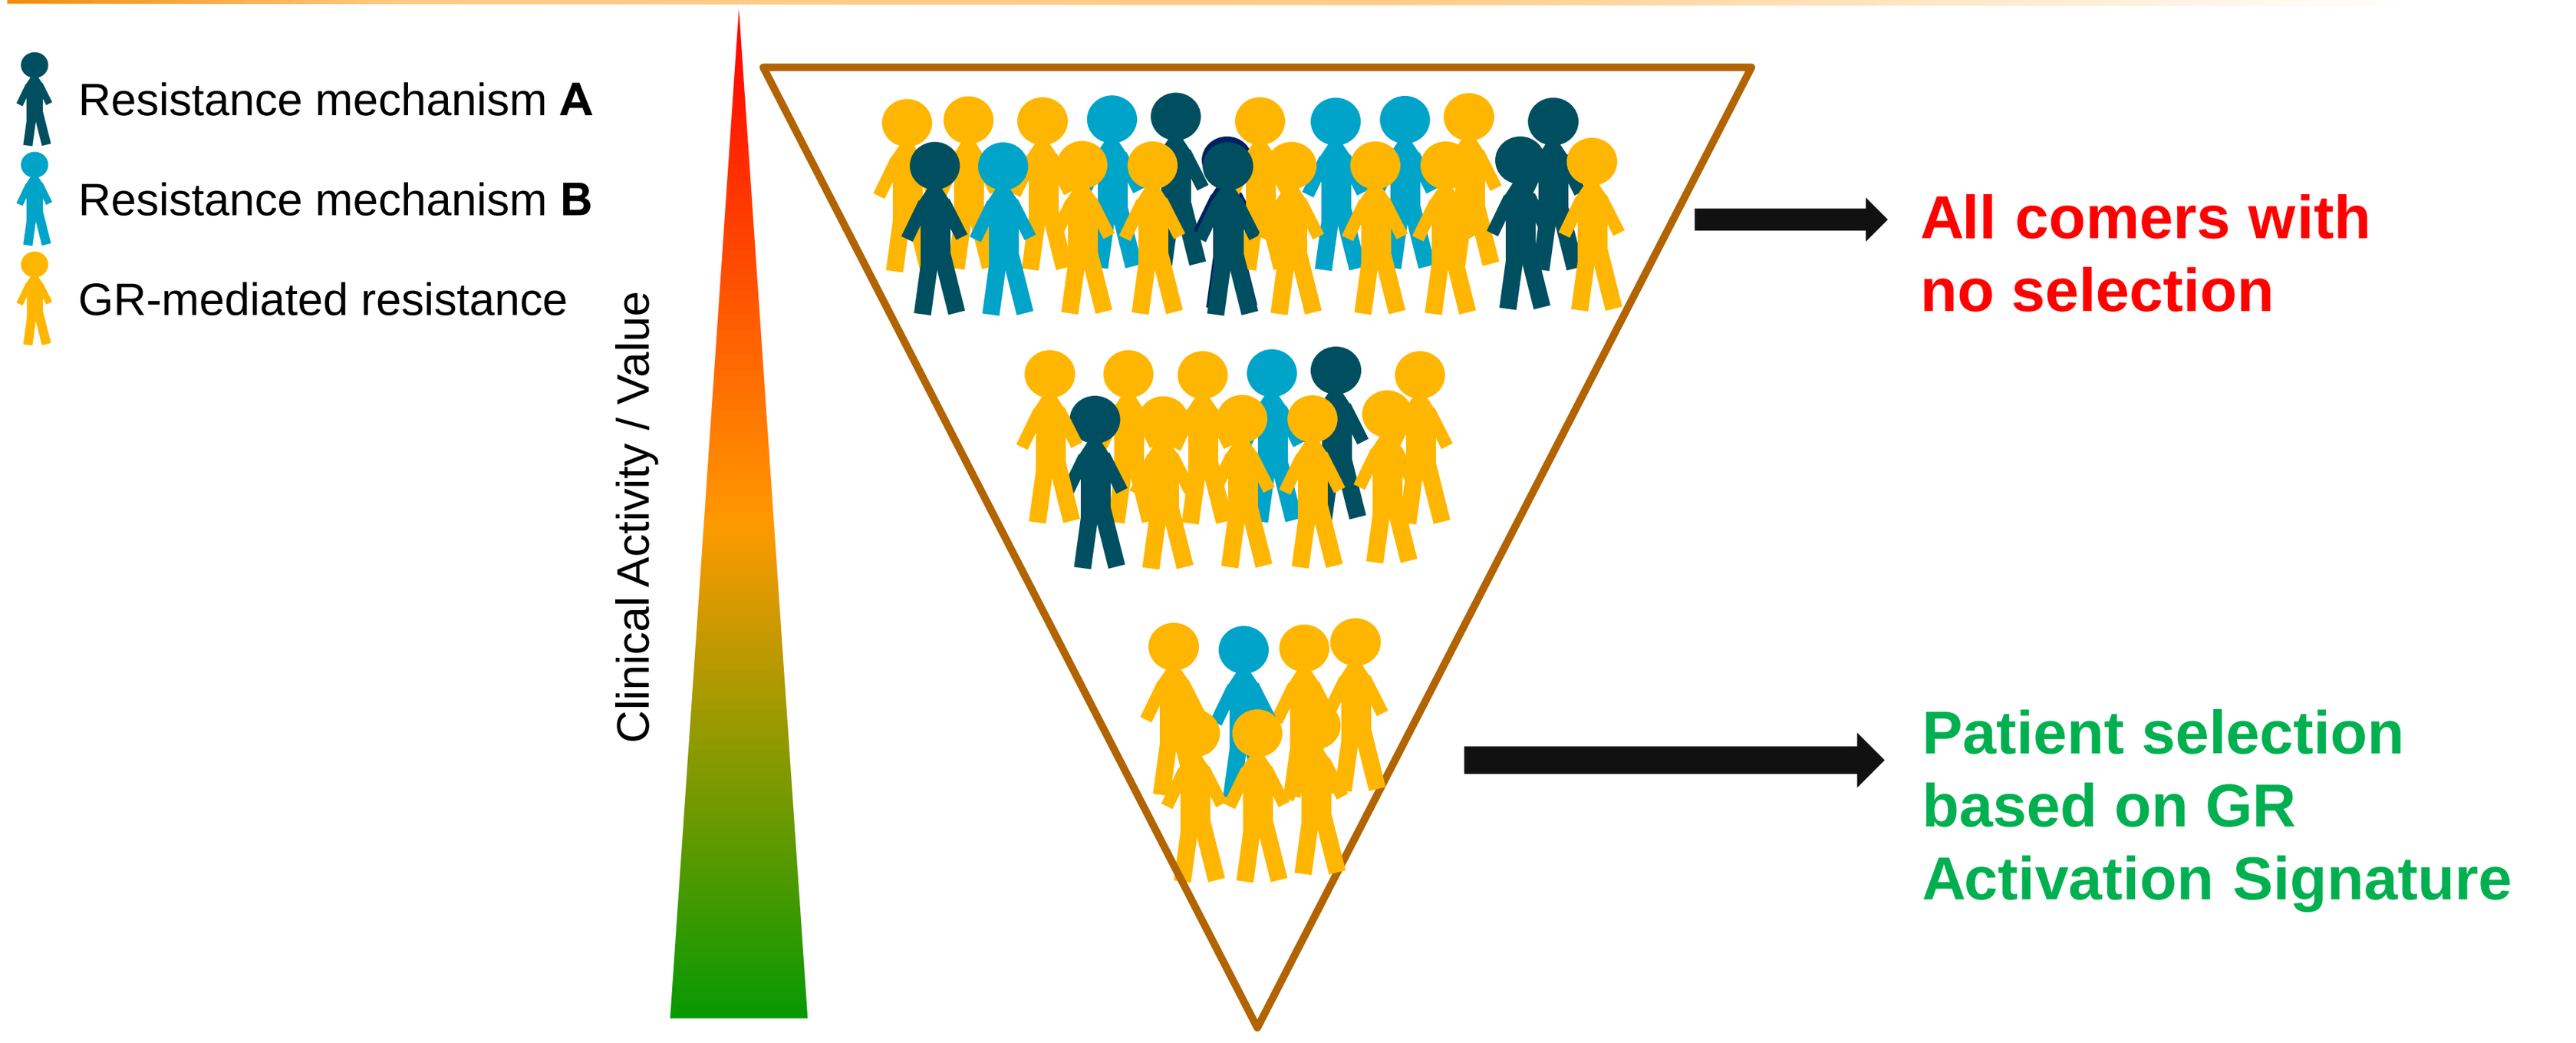
Resistance mechanism A
Resistance mechanism B
GR-mediated resistance
Clinical Activity / Value
All comers with no selection
Patient selection based on GR Activation Signature

Patients with activated GR signaling and GR-driven chemoresistance may benefit from ORIC-101

4. GR Activation Signature derived from 8 TNBC, 12 PDAC, and 12 NSCLC cell lines

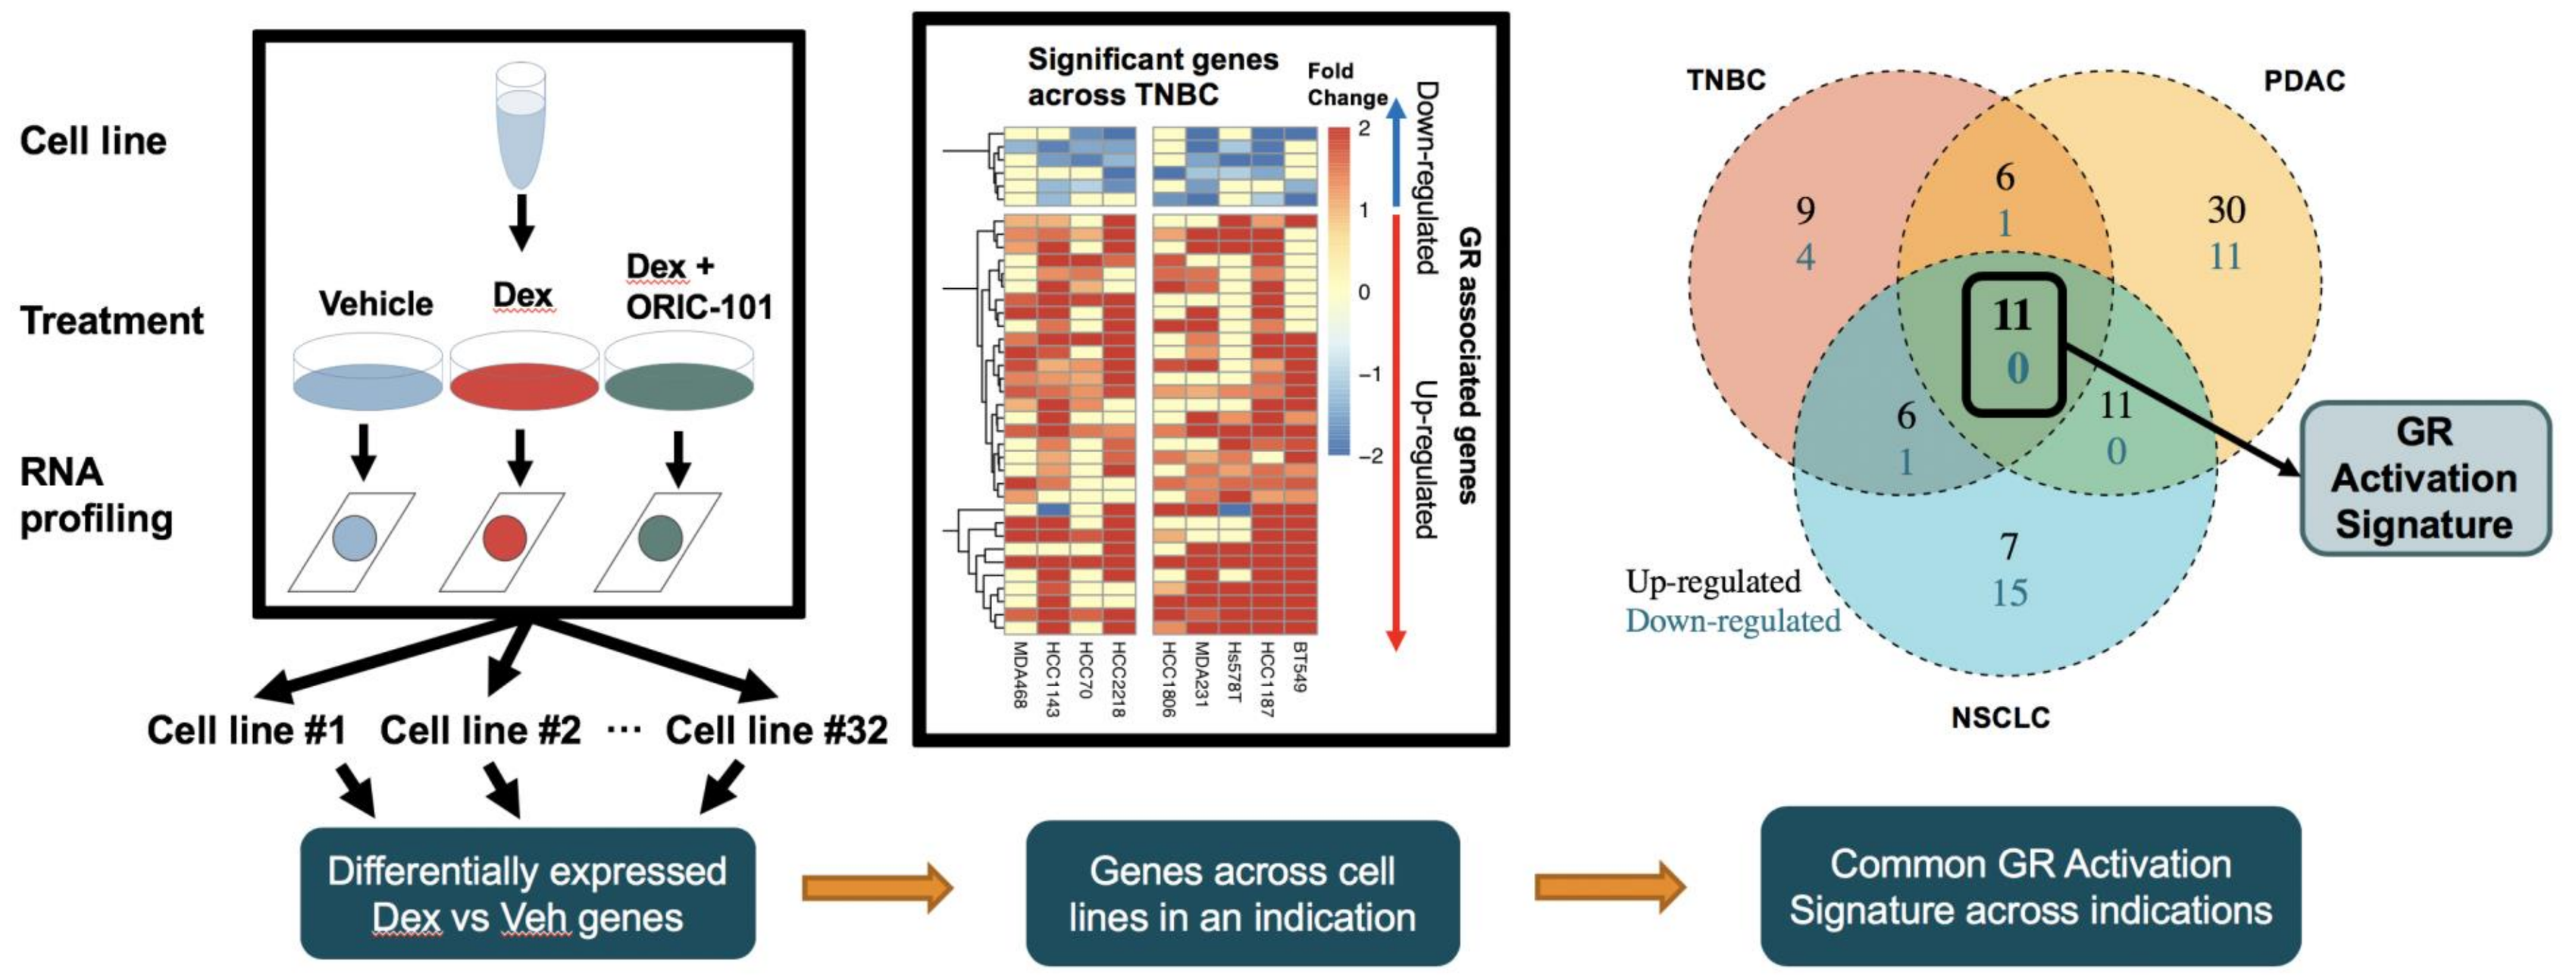
Cell line
Treatment
RNA profiling
Vehicle
Dex
Dex + ORIC-101
Cell line #1
Cell line #2
Cell line #32
Significant genes across TNBC
Fold Change
Down-regulated
Up-regulated
GR associated genes
TNBC
PDAC
NSCLC
Up-regulated
Down-regulated
GR Activation Signature
Differentially expressed Dex vs Veh genes
Genes across cell lines in an indication
Common GR Activation Signature across indications

GR Activation Signature is identified across a wide range of cancer models

5. Cell lines show common and specific pathways modulated by GR activation

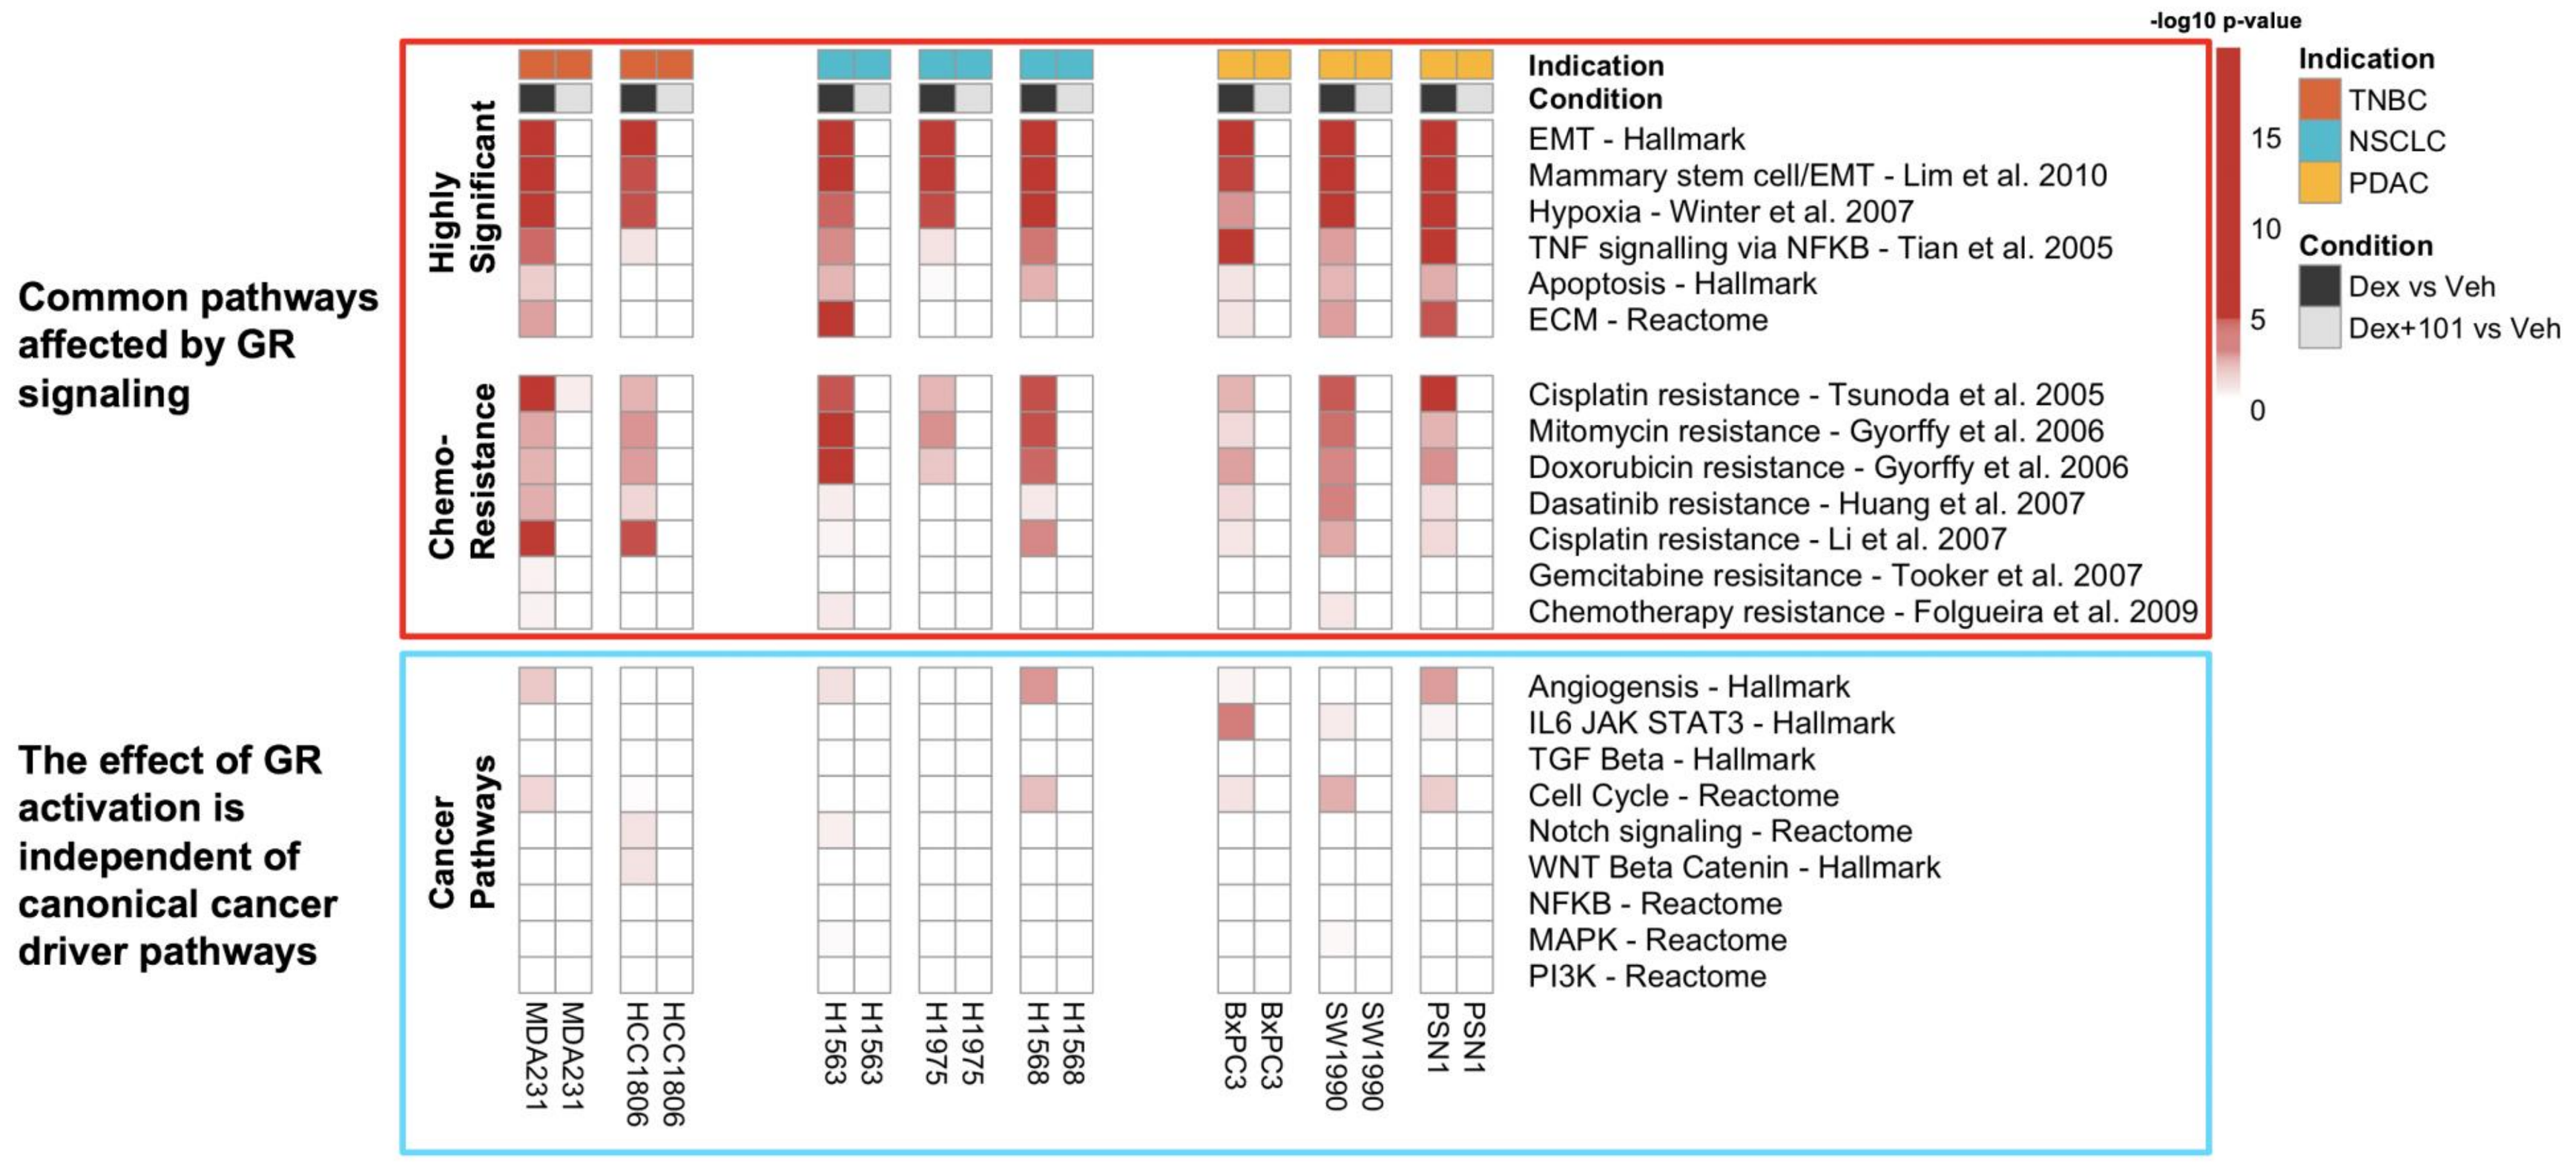
Common pathways affected by GR signaling
The effect of GR activation is independent of canonical cancer driver pathways
Highly Significant
Chemo-Resistance
Cancer Pathways
Indication
Condition
EMT - Hallmark
Mammary stem cell/EMT - Lim et al. 2010
Hypoxia - Winter et al. 2007
TNF signalling via NFKB - Tian et al. 2005
Apoptosis - Hallmark
ECM - Reactome
Cisplatin resistance - Tsunoda et al. 2005
Mitomycin resistance - Gyorffy et al. 2006
Doxorubicin resistance - Gyorffy et al. 2006
Dasatinib resistance - Huang et al. 2007
Cisplatin resistance - Li et al. 2007
Gemcitabine resisitance - Tooker et al. 2007
Chemotherapy resistance - Folgueira et al. 2009
Angiogensis - Hallmark
IL6 JAK STAT3 - Hallmark
TGF Beta - Hallmark
Cell Cycle - Reactome
Notch signaling - Reactome
WNT Beta Catenin - Hallmark
NFKB - Reactome
MAPK - Reactome
PI3K - Reactome
-log10 p-value
TNBC
NSCLC
PDAC
Dex vs Veh
Dex+101 vs Veh
MDA231
HCC1806
H1563
H1975
H1568
BxPC3
SW1990
PSN1

ORIC-101 inhibits pathways modulated by GR activation

6. Association of the GR Activation Signature with pathways in human TCGA tumors

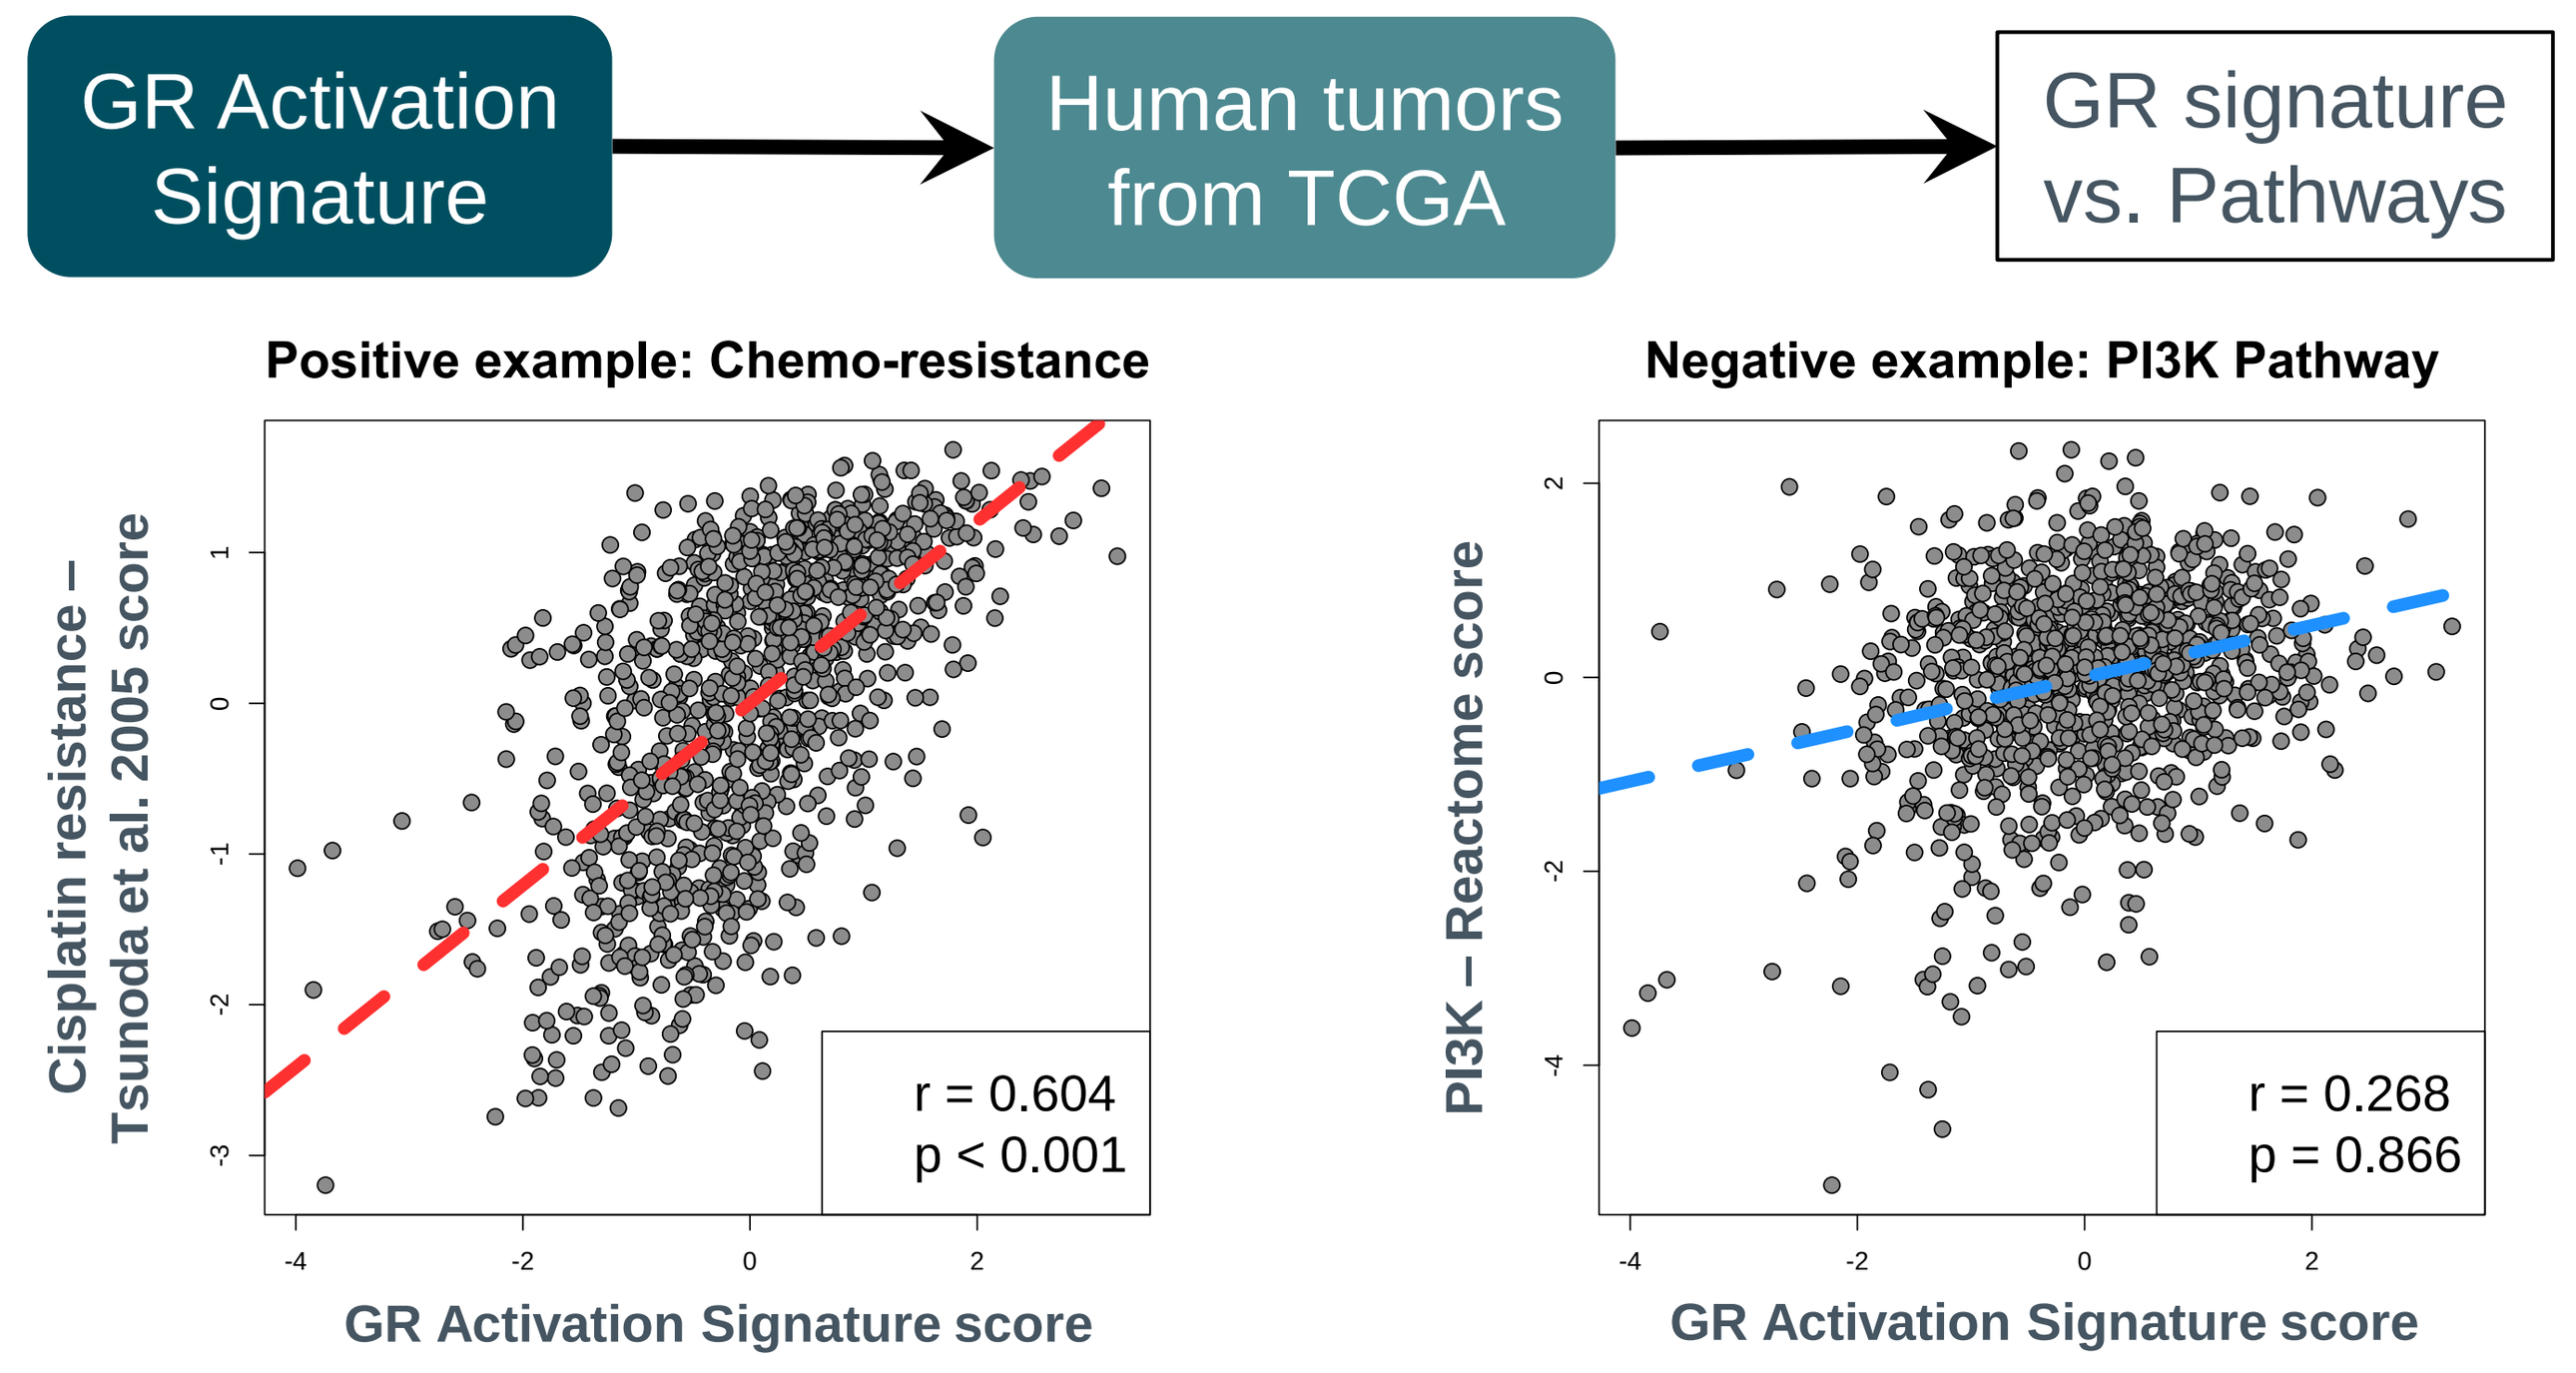
GR Activation Signature
Human tumors from TCGA
GR signature vs. Pathways
Positive example: Chemo-resistance
Negative example: PI3K Pathway
Cisplatin resistance – Tsunoda et al. 2005 score
PI3K – Reactome score
r = 0.604
p < 0.001
r = 0.268
p = 0.866
GR Activation Signature score

7. GR activation captures the same biological processes in human tumors

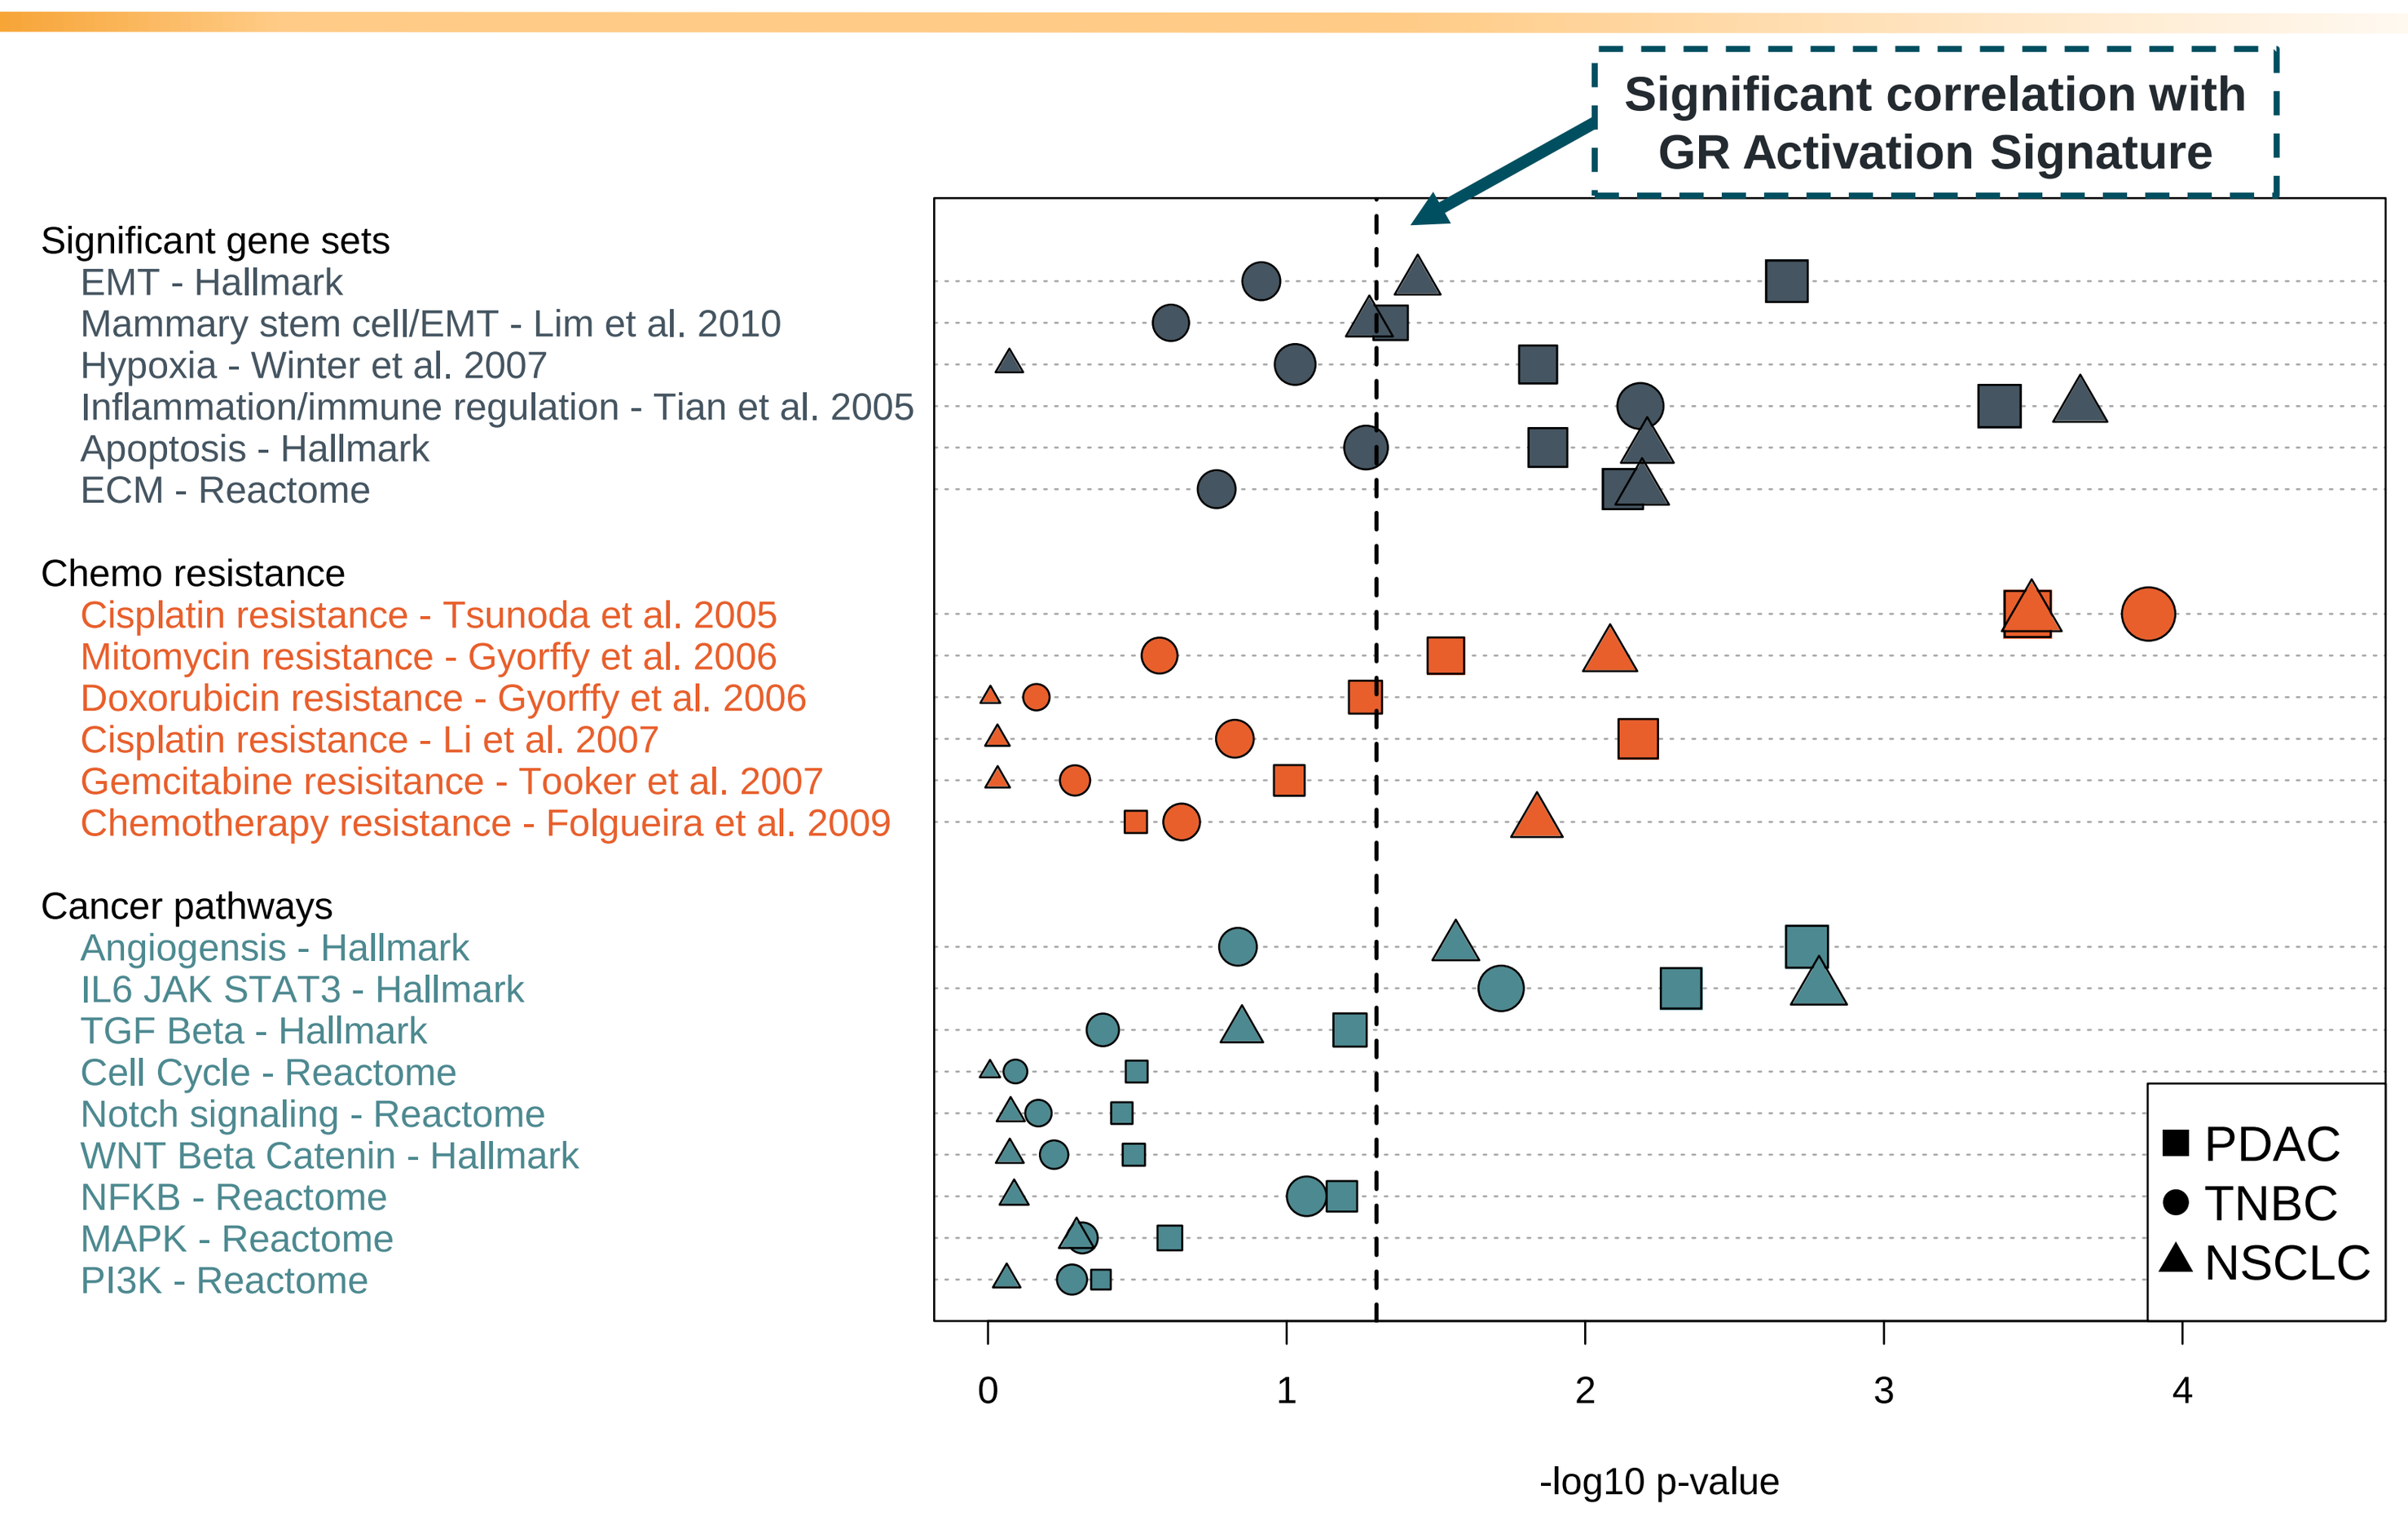
Significant correlation with GR Activation Signature
Significant gene sets
EMT - Hallmark
Mammary stem cell/EMT - Lim et al. 2010
Hypoxia - Winter et al. 2007
Inflammation/immune regulation - Tian et al. 2005
Apoptosis - Hallmark
ECM - Reactome
Chemo resistance
Cisplatin resistance - Tsunoda et al. 2005
Mitomycin resistance - Gyorffy et al. 2006
Doxorubicin resistance - Gyorffy et al. 2006
Cisplatin resistance - Li et al. 2007
Gemcitabine resisitance - Tooker et al. 2007
Chemotherapy resistance - Folgueira et al. 2009
Cancer pathways
Angiogensis - Hallmark
IL6 JAK STAT3 - Hallmark
TGF Beta - Hallmark
Cell Cycle - Reactome
Notch signaling - Reactome
WNT Beta Catenin - Hallmark
NFKB - Reactome
MAPK - Reactome
PI3K - Reactome
PDAC
TNBC
NSCLC
-log10 p-value

Pathways associated with the GR Activation Signature in cell lines are recapitulated in human tumors from TCGA

CONCLUSIONS

- ORIC-101 reverses GR-activated transcriptional programs at both the gene and pathway level in preclinical models.
- GR Activation Signature translates from cell lines to human tumors.
- The GR Activation Signature allows for monitoring target engagement of ORIC-101 and patient selection based on activated GR signaling.
- ORIC-101 is in early clinical development in combination with nab-paclitaxel in solid tumors (NCT03928314) and in combination with enzalutamide in metastatic prostate cancer (NCT04033328) where the GR activation signature is being evaluated.

Acknowledgements: Jessica D Sun, Melissa R Junttila, Ryan Athearn, Edna Chow Maneval, Pratik S Multani, and Jacob Chacko

AACR Virtual Annual Meeting II, June 22-24, 2020, Poster #4120